Poster F150 5th International Congress on Drug Therapy in HIV Infection, Glasgow, UK (October 22-26, 2000)

Kaletra (ABT-378/ritonavir) and Efavirenz: 48-Week Safety/Efficacy Evaluation in Multiple PI-Experienced Patients

J. Rockstroh[1], S. Brun[12], R. Bertz[12], S. Chen[12], N. Clumeck[2], A. Lazzarin[3], P. M. Girard[4], S. Becker[5], A. Telenti[6], F. Bergmann[7], S. Danner[8], D. Ho[9], R. Tubiana[10], G. Carosi[11], J. Sylte[12], A. Hsu[12], D. Kempf[12] and E. Sun[12] for the M98-957 Study Group.
Med. Klinik I der Universitat Bonn[1], H.U. Saint-Pierre[2], Osp. S. Raffaele[3], H. Rotschild[4], Pacific Horizons[5], H. de Beaumont[6], Charite Campus Virchow[7], Academic Med. Ctr. Amsterdam[8], Aaron Diamond[9], H. Pitie-Salpetriere[10], U. DiBrescia[11] and Abbott Laboratories[12]

BACKGROUND

ABT-378 (lopinavir) is a novel HIV protease inhibitor (PI) that is co-formulated with ritonavir, an inhibitor of cytochrome P450 3A. It is exquisitely sensitive to pharmacokinetic enhancement by ritonavir, resulting in substantially increased ABT-378 drug exposure, even at low ritonavir doses. At the dosage selected for phase III clinical trials, 400 mg ABT-378/100 mg ritonavir BID (as 3 co-formulated capsules BID), ritonavir concentrations are below those required for antiviral activity.[1] The mean ABT-378 C_{trough}/EC_{50} ratio (Inhibitory Quotient or IQ) for wild-type HIV is ≥75 when dosed at 400/100 mg BID, potentially providing a pharmacologic barrier to the emergence of viral resistance and activity against resistant virus.[1] In contrast, the IQs for currently available PIs, whether dosed as single PIs or in conjunction with ritonavir, range from 1-26 (Figure 1), based on the EC_{50} values determined in the presence of 50% human serum.[2] C_{trough} values in HIV+ patients for currently available PIs are derived from published sources including package inserts.[3-14]

The efficacy and safety of Kaletra (lopinavir/ritonavir, formerly known as ABT-378/r) are currently being studied in HIV-infected patients, both antiretroviral-naïve and PI-experienced. At 96 weeks, HIV viral load is <400 copies/mL in 97% of antiretroviral-naïve patients on treatment (intent-to-treat missing=failure:83%) and <50 copies/mL in 92% of patients on treatment (ITT M=F:78%) receiving Kaletra 400/100 mg BID with d4T/3TC in Study M97-720.[15] Results from a previous Phase I parallel arm pharmacokinetic drug interaction study indicated a small effect of Kaletra on efavirenz pharmacokinetics that does not require a dosage adjustment of efavirenz. However, due to a high subject discontinuation rate in this study secondary to rash, the sample size was inadequate to characterize the effect of efavirenz on ABT-378 pharmacokinetic parameters.

The M98-957 study is an ongoing phase II, open-label, randomized trial of Kaletra, in combination with efavirenz, in multiple PI-experienced/NNRTI-naïve patients. Pharmacokinetic data and 48 week efficacy and safety data are reported here. Association of virologic response at Week 24 with baseline virologic genotype/phenotype is presented as well.

Figure 1. C_{trough}/EC_{50} Ratio (Inhibitory Quotient) for PIs Given Alone or with RTV*

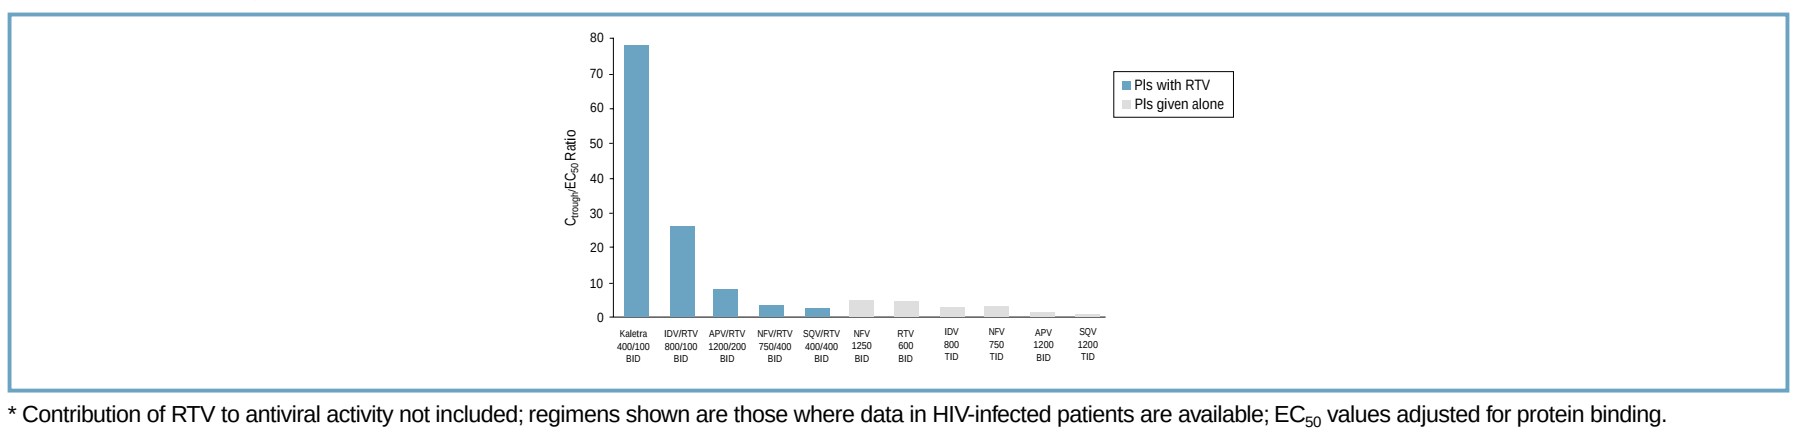
C_{trough}/EC_{50} Ratio
0
10
20
30
40
50
60
70
80
PIs with RTV
PIs given alone
Kaletra 400/100 BID
IDV/RTV 800/100 BID
APV/RTV 1200/200 BID
NFV/RTV 750/400 BID
SQV/RTV 400/400 BID
NFV 1250 BID
RTV 600 BID
IDV 800 TID
NFV 750 TID
APV 1200 BID
SQV 1200 TID

* Contribution of RTV to antiviral activity not included; regimens shown are those where data in HIV-infected patients are available; EC_{50} values adjusted for protein binding.

METHODS

Entry Criteria

- HIV RNA >1,000 copies/mL on present PI regimen (no CD4 cell count restriction).
- On treatment with at least 1 PI for ≥ 8 weeks at study entry.
- Multiple PI-experienced (history of sequential or concurrent treatment with at least 2 PIs for at least 3 months each).
- NNRTI-naïve.

Study Design and Analysis

- 57 patients received Kaletra 400/100 mg (3 coformulated capsules) BID in place of their current PI, in combination with efavirenz (600 mg QD) and NRTIs as determined by the investigator, for the first 13 days of the study (Figure 2).
- On study Day 14, patients randomized to Arm B (n=28) increased their Kaletra dose to 533/133 mg (4 coformulated capsules) BID, while patients in Arm A (n=29) remained on the 400/100 mg BID dose (Figure 2).
- ABT-378 trough levels were drawn at Week 2; full PK was performed at Week 5; efavirenz levels were also drawn at Weeks 2 and 5.
- Plasma HIV RNA was quantified using the Roche Amplicor HIV-1 Monitor (assay lower limit of quantitation [LLQ] 400 copies/mL).
- All patients in Arm A began conversion to the 533/133 BID dose after Week 24 and completed the process prior to their Week 48 visit.

Figure 2. M98-957 Study Design

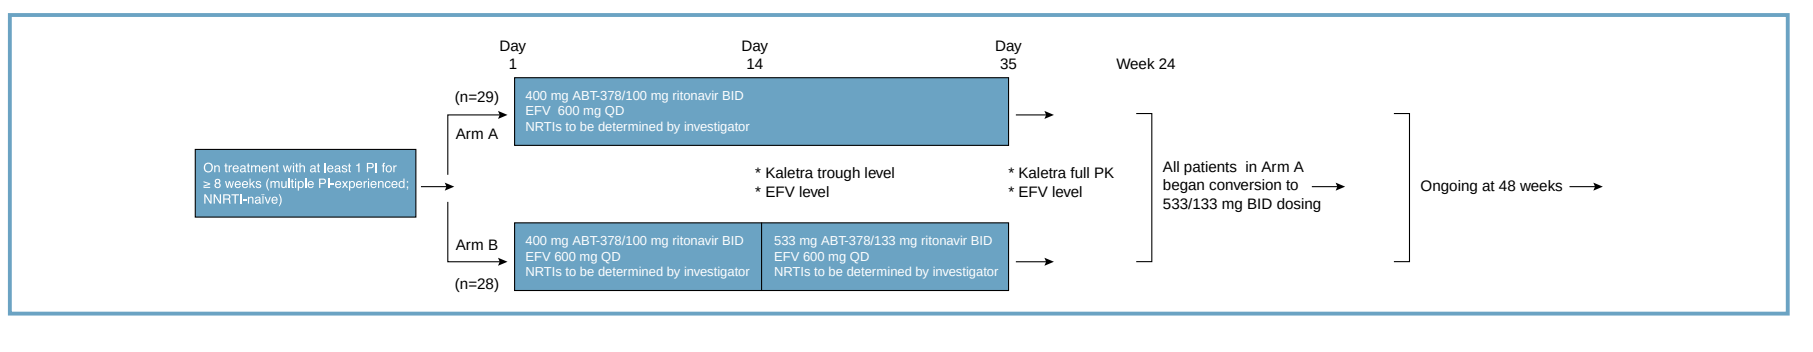
Day 1
Day 14
Day 35
Week 24
(n=29)
Arm A
400 mg ABT-378/100 mg ritonavir BID
EFV 600 mg QD
NRTIs to be determined by investigator
On treatment with at least 1 PI for ≥ 8 weeks (multiple PI-experienced; NNRTI-naïve)
* Kaletra trough level
* EFV level
* Kaletra full PK
* EFV level
All patients in Arm A began conversion to 533/133 mg BID dosing
Ongoing at 48 weeks
Arm B
(n=28)
400 mg ABT-378/100 mg ritonavir BID
EFV 600 mg QD
NRTIs to be determined by investigator
533 mg ABT-378/133 mg ritonavir BID
EFV 600 mg QD
NRTIs to be determined by investigator

RESULTS

Table 1. Baseline Characteristics

	400/100 mg (n=29)	533/133 mg (n=28)
Mean age, years	41	43
Men, %	76%	82%
Race or ethnicity, %		
Caucasian	86%	89%
Black	7%	11%
Hispanic	14%	0%
Asian	0%	0%
HIV RNA level (log_{10} copies/mL)		
Median	4.7	4.2
Range	2.6-6.4	3.2-6.1
CD4 count (cells/mm³)		
Median	218	271
Range	26-853	13-1,030

Pre-Study Antiretroviral Therapy

- Mean number of prior ARVs=7 (range: 3-10).
- Mean number of prior PIs=3 (range: 1-4); for patients who received RTV pre-study, 66% (29/44) received it as dual PI therapy.
- Mean number NRTIs at baseline=2 (range: 1-4).
- 75% of patients did not receive a new NRTI in conjunction with Kaletra and efavirenz within the first eight weeks of study.

Table 2. Pre-Study Antiretroviral Therapy

	% Patients	
Prior ARV	400 mg/100 mg (n=29)	533 mg/133 mg (n=28)
Indinavir	83%	89%
Nelfinavir	55%	61%
Ritonavir	90%	64%
Saquinavir	69%	75%
Abacavir	14%	21%
Didanosine	83%	75%
Lamivudine	93%	89%
Stavudine	83%	100%
Zalcitabine	45%	46%
Zidovudine	97%	89%
Hydroxyurea	7%	18%

Baseline Viral Susceptibility

- Protease inhibitor phenotypic susceptibility data were available for 56 of 57 baseline viral isolates (ViroLogic, Inc. method).
- Sixty-eight percent (38/56) of patients had baseline viral isolates demonstrating cross resistance (≥4-fold increase in EC_{50} relative to wild-type virus) to at least 3 licensed PIs.
- Forty-three percent of baseline isolates (24/56) demonstrated ≥10-fold increase in EC_{50} of ABT-378 relative to wild-type virus.

Table 3. Baseline Phenotypic Susceptibility

	Median Fold Change in EC_{50} Relative to Wild-Type Virus (Range)	
Protease Inhibitor	400 mg/100 mg (n=28)	533 mg/133 mg (n=28)
ABT-378	9 (0.5 – 96.0)	4 (0.7 – 67.0)
Ritonavir	37 (0.8 – 315.8)	19 (0.5 – 148.0)
Indinavir	11 (0.8 – 171.4)	9 (0.9 – 73.0)
Nelfinavir	15 (1.1 – 158.0)	23 (1.1 – 131.0)
Amprenavir	2 (0.5 – 49.0)	3 (0.6 – 29.0)
Saquinavir	6 (0.4 – 461.5)	5 (0.4 – 545.5)

Pharmacokinetic Data

- ABT-378 levels achieved with the 400/100 mg dose are reduced when co-dosed with efavirenz (C_{trough} reduced ~33%; AUC reduced ~25%).
- Kaletra 533/133 mg dose with efavirenz provides similar ABT-378 exposures to the 400/100 mg dose without efavirenz based on historical controls.[1]
- Efavirenz levels are similar for both Kaletra dose levels studied.

Patient Disposition at Week 48

	Dose Groups	
	400/100 mg	533/133 mg
Patients Enrolled	29	28
Patients Discontinued at or Before Week 48	7	4
Discontinuations Related to Kaletra*		
CNS sx	1	1
Lactic Acidosis		1
Other Reasons for Discontinuation		
Adverse Event (CNS sx, hypertriglyceridemia)	2	0
Virologic Failure (Investigator Opinion)	3	1
Personal Reasons	1	0
Death (Septic Shock)	0	1

*Adverse events noted as probably or possibly related to Kaletra are included.

Viral Load Suppression at 48 Weeks

- On-treatment analysis (OT): 77% of patients (17/22) in the 400/100 mg dose arm and 83% of patients (20/24) in the 533/133 mg dose arm had viral load (VL) <400 copies/mL at Week 48 (Figure 3).
- Intent-to-treat analysis (ITT M=F; missing values [M] considered as treatment failure [F]): 59% of patients (17/29) in the 400/100 mg dose arm and 71% of patients (20/28) in the 533/133 mg dose arm had VL <400 copies/mL at Week 48 (Figure 4).

Figure 3. M98-957 HIV RNA <400 Copies/mL (OT)

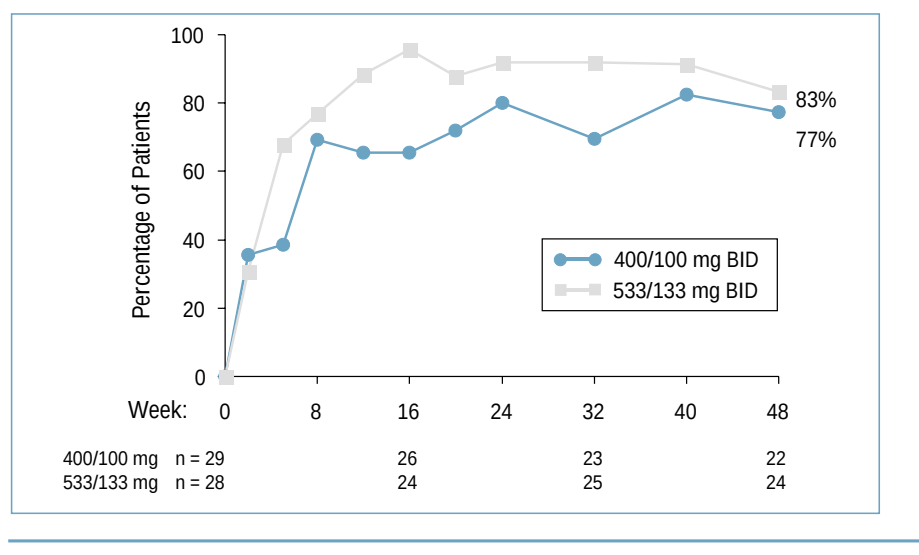
Percentage of Patients
100
80
60
40
20
0
Week: 0 8 16 24 32 40 48
83%
77%
400/100 mg BID
533/133 mg BID
400/100 mg n = 29 26 23 22
533/133 mg n = 28 24 25 24

Figure 4. M98-957 HIV RNA <400 Copies/mL (ITT M=F)

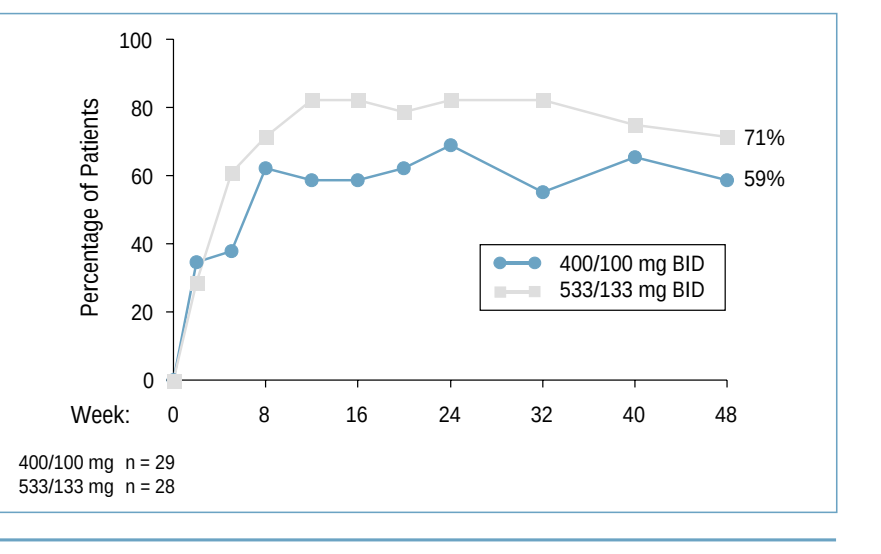
Percentage of Patients
100
80
60
40
20
0
Week: 0 8 16 24 32 40 48
71%
59%
400/100 mg BID
533/133 mg BID
400/100 mg n = 29
533/133 mg n = 28

Association of Week 24 Virologic Response with Baseline Virologic Genotype/Phenotype

Figure 5. M98-957: Week 24 Virologic Response with Respect to Baseline Phenotype

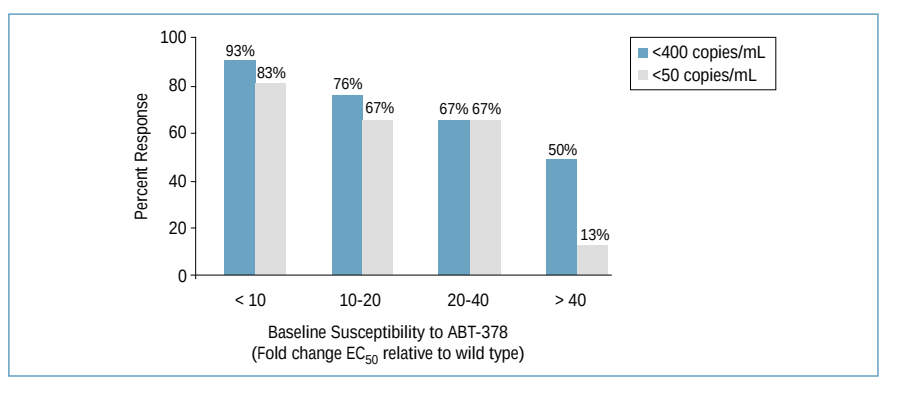
Percent Response
<400 copies/mL
<50 copies/mL
93%
83%
76%
67%
67% 67%
50%
13%
< 10
10-20
20-40
> 40
Baseline Susceptibility to ABT-378
(Fold change EC_{50} relative to wild type)

Figure 6. M98-957: Week 24 Virologic Response with Respect to Baseline Genotype

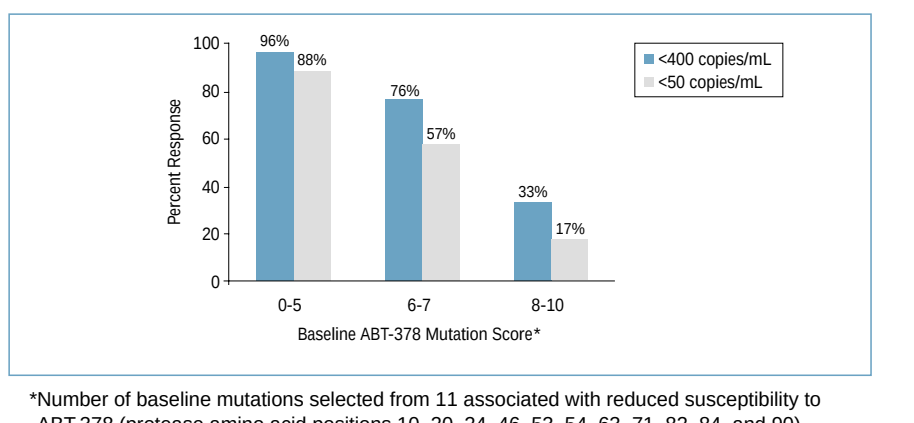
Percent Response
<400 copies/mL
<50 copies/mL
96%
88%
76%
57%
33%
17%
0-5
6-7
8-10
Baseline ABT-378 Mutation Score*

*Number of baseline mutations selected from 11 associated with reduced susceptibility to ABT-378 (protease amino acid positions 10, 20, 24, 46, 53, 54, 63, 71, 82, 84, and 90)

- At 24 weeks high response rates at both <400 copies/mL (93%) and <50 copies mL (83%) were observed in patients whose baseline isolates displayed <10-fold reduced *in vitro* susceptibility to ABT-378 at baseline (Figure 5). Response rates diminished incrementally in patients with 10-20, 20-40, and >40-fold reduced susceptibility to ABT-378 at baseline.
- Similarly, very high response rates at 24 weeks at both <400 copies/mL (96%) and <50 copies/mL (88%) were observed in patients whose baseline isolates contained 0-5 protease mutations associated with reduced *in vitro* susceptibility to ABT-378 (Figure 6). Response rates diminished incrementally in patients with a baseline ABT-378 mutation score of 6-7 and 8-10.

CD4 Response at 48 Weeks

- Among patients on study at Week 48, the mean CD4 cell count was 359 cells/μL for the 400/100 mg dose arm and 432 cells/μL for the 533/133 dose arm (mean increases from baseline of 116 and 67 cells/μL in the 400/100 and 533/133 mg dose arms, respectively) (Figure 7). No significant differences between treatment groups were observed after Week 2.

Figure 7. M98-957: Mean Change from Baseline in CD4 Cell Count

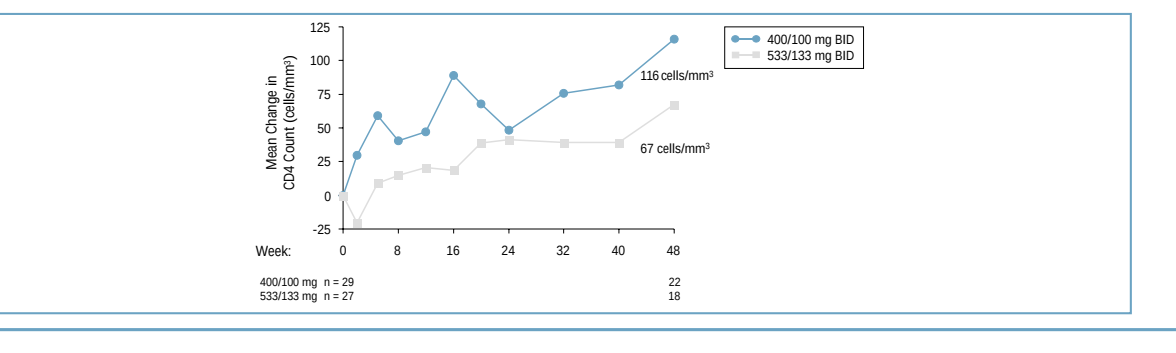
Mean Change in CD4 Count (cells/mm³)
125
100
75
50
25
0
-25
400/100 mg BID
533/133 mg BID
116 cells/mm³
67 cells/mm³
Week: 0 8 16 24 32 40 48
400/100 mg n = 29 22
533/133 mg n = 27 18

Safety and Tolerability

- The most common study drug related adverse events of at least moderate severity were diarrhea and asthenia, while the most common laboratory abnormality was lipid elevations (Table 4). Of note, lipid measurements were made without regard to fasting.
- Eleven patients discontinued the study through 48 weeks; of these, 3 discontinuations were due to adverse events/laboratory abnormalities probably or possibly related to Kaletra in the opinion of the investigator.
- None of the patients who experienced amylase values >2 x ULN through Week 48 had associated elevations of pancreatic amylase (>2 x ULN).

Table 4. Most Common Adverse Events* and Grade 3/4 Laboratory Abnormalities

	% Patients	
	400 mg/100 mg (n=29)	533 mg/133 mg (n=28)
Diarrhea	10%	14%
Asthenia	7%	14%
Glucose (>250 mg/dL)	10%	0%
SGOT/AST (>5 x ULN)	0%	7%
SGPT/ALT (>5 x ULN)	0%	7%
Total Cholesterol (>300 mg/dL)	35%	46%
Triglycerides (>750 mg/dL)	41%	39%
Amylase (>2 x ULN)	3%	14%
Neutrophils (<0.75 x 10^9/L)	7%	7%

*Adverse events of at least moderate severity and probable, possible, or unknown relationship to Kaletra included.

CONCLUSIONS

- Kaletra and efavirenz exhibit a potent antiviral effect through 48 weeks of therapy: Viral load was <400 copies/mL in 77% (Arm A) and 83% (Arm B) of multiple PI-experienced patients on treatment (ITT M=F: 59% and 71%, respectively).
- Three out of 57 patients discontinued therapy due to adverse events considered probably or possibly related to Kaletra at 48 weeks.
- Through 24 weeks, Kaletra and efavirenz demonstrated significant activity against HIV strains with up to 40-fold reduced susceptibility to ABT-378 or up to 7 mutations associated with ABT-378 resistance.
- While ABT-378 AUC and trough levels are reduced 25% and 33% when the 400/100 mg dose level of Kaletra is coadministered with efavirenz, the 533/133 mg dose with efavirenz provides similar ABT-378 exposures to the 400/100 mg dose without efavirenz based on historical controls.
- Efavirenz levels are similar between the Kaletra dose levels studied.
- Kaletra should be increased to 533/133 mg BID when coadministered with efavirenz in patients where reduced susceptibility to ABT-378 is suspected by treatment history or laboratory evidence.

ACKNOWLEDGMENTS

Efavirenz was supplied by DuPont Pharmaceuticals Company.
Phenotype testing of baseline viral isolates was performed by ViroLogic, Inc.

REFERENCES

1. Bertz R, Lam W, Brun S, et al. Multiple-dose pharmacokinetics (PK) of ABT-378/ritonavir (ABT-378/r) in HIV+ subjects. 39th Interscience Conference on Antimicrobial Agents and Chemotherapy, San Francisco, USA, 1999 (abstract 0327).
2. Kempf DJ, Molla A, Hsu A. Protease inhibitors as anti-HIV agents. Antiretroviral Therapy, American Society for Microbiology Press, E DeClerq, editor. In press.
3. Ritonavir Package Insert.
4. Indinavir Package Insert.
5. Amprenavir Package insert.
6. Nelfinavir Package Insert.
7. Saquinavir (Fortavase) Package Insert.
8. Cohen C, et al. Potent and convenient Fortovase™ (SQV) SGC BID regimens in combination with 2 nucleosides or nelfinavir (NFV) plus 1 nucleoside in HIV-1 infected patients. 12th World AIDS Conference, Geneva, Switzerland, 1998, (abstract 12314).
9. Saag M, et al. Saquinavir systemic exposure and safety on once daily administration of Fortovase™ (Saquinavir) soft gel capsules (FTV) in combination with low-dose ritonavir. 39th Interscience Conference on Antimicrobial Agents and Chemotherapy, San Francisco, USA, 1999, (abstract 330).
10. Shulman N, et al. Ritonavir intensification in indinavir recipients with detectable HIV RNA levels. 7th Conference on Retroviruses and Opportunistic Infections, San Francisco, USA (abstract 534).
11. Piscitelli S, et al. The addition of a second protease inhibitor eliminates Amprenavir-Efavirenz drug interactions and increases plasma Amprenavir concentrations. 7th Conference on Retroviruses and Opportunistic Infections, San Francisco, USA (abstract 78).
12. Mallolas J, et al. A dose-finding study of once-daily Indinavir/Ritonavir plus Combivir® in HIV infected patients. First International Workshop on Clinical Pharmacology of HIV Therapy, March 2000 (abstract 2.14).
13. Flexner C, et al. Steady-state pharmacokinetic interactions between ritonavir (RTV), nelfinavir (NFV), and the nelfinavir active metabolite M8 (AG1402), 12th World AIDS Conference, Geneva (abstract 42265).
14. Cameron DW, et al. Ritonavir and saquinavir combination therapy for the treatment of HIV infection. AIDS, 1999, 12(2) 213-224.
15. Benson C, King M, Brun S, et al. ABT-378/ritonavir (ABT-378/r) in antiretroviral – naïve HIV+ patients: 96 weeks. 40th Interscience Conference on Antimicrobial Agents and Chemotherapy, Toronto, Canada, 2000 (abstract 546).

Kaletra™ is not yet licensed in the U.K.

E002404C